Utilizing the Electronic Trial Master File for Clinical Studies to Support FDA Tobacco Product Applications

Jeffrey Coffield, Claude Cavallo, Heather Green and Ken Szeliga
Scientific & Regulatory Affairs, RAI Services Company, Winston-Salem, NC, USA

Introduction

Most new tobacco products introduced to the US market require the submission of a Premarket Tobacco Product (PMTA) application to the FDA Center for Tobacco Products and receive marketing authorization. This application comprises data generated across multiple scientific disciplines to collectively demonstrate that the new product is appropriate for the protection of public health. Clinical trial data is an important aspect of this scientific evidence, as it directly represents potential risks and benefits of new tobacco product use in humans.

Clinical studies are often lengthy endeavors with potentially large numbers of subjects and study conditions conducted across multiple study sites. These studies produce vast quantities of documentation including every record associated with each individual subject's trial participation. This information must be tracked, verified, stored, and easily accessible upon request, as per FDA regulatory requirements. An electronic Trial Master File (eTMF) system is a secure database that ensures all records associated with the clinical trial are managed in compliance with FDA regulations promulgated under good clinical practice (GCP), HIPPA and FDA 21 CFR Part 11.

Trial Master File Importance

Why: Regulatory Requirement, Tells Story

What: Essential Documents (ICH GCP Sec 8)

Who: Sponsors, Sites, CROs, Vendors, IRB

Where: Source, Format, TMF Index (TOC)

When: Milestones, Events: Before, During and After trial closure

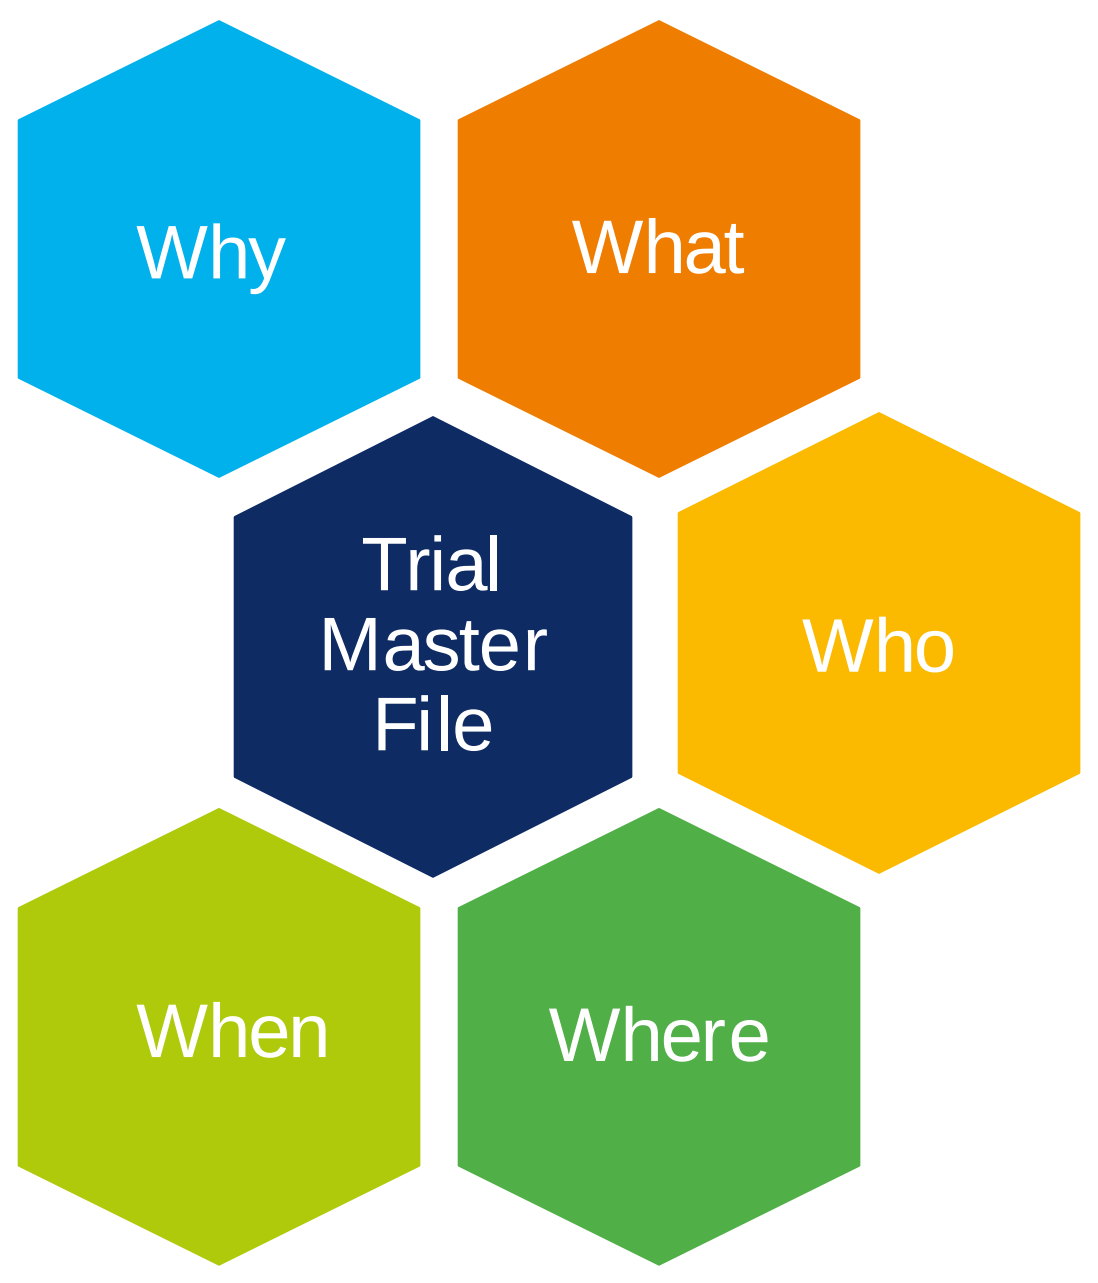
Why
What
Trial Master File
Who
When
Where

Trial Master File Importance

Best practices related to submitting documents into the eTMF in a timely fashion, evaluating the quality of the documents, and reconciling the eTMF for completeness are key to quality documentation. If discovered, the details surrounding issues should be documented to provide rationale for handling concerns exhibiting oversight and resolution of those issues.

TMF Reference Model

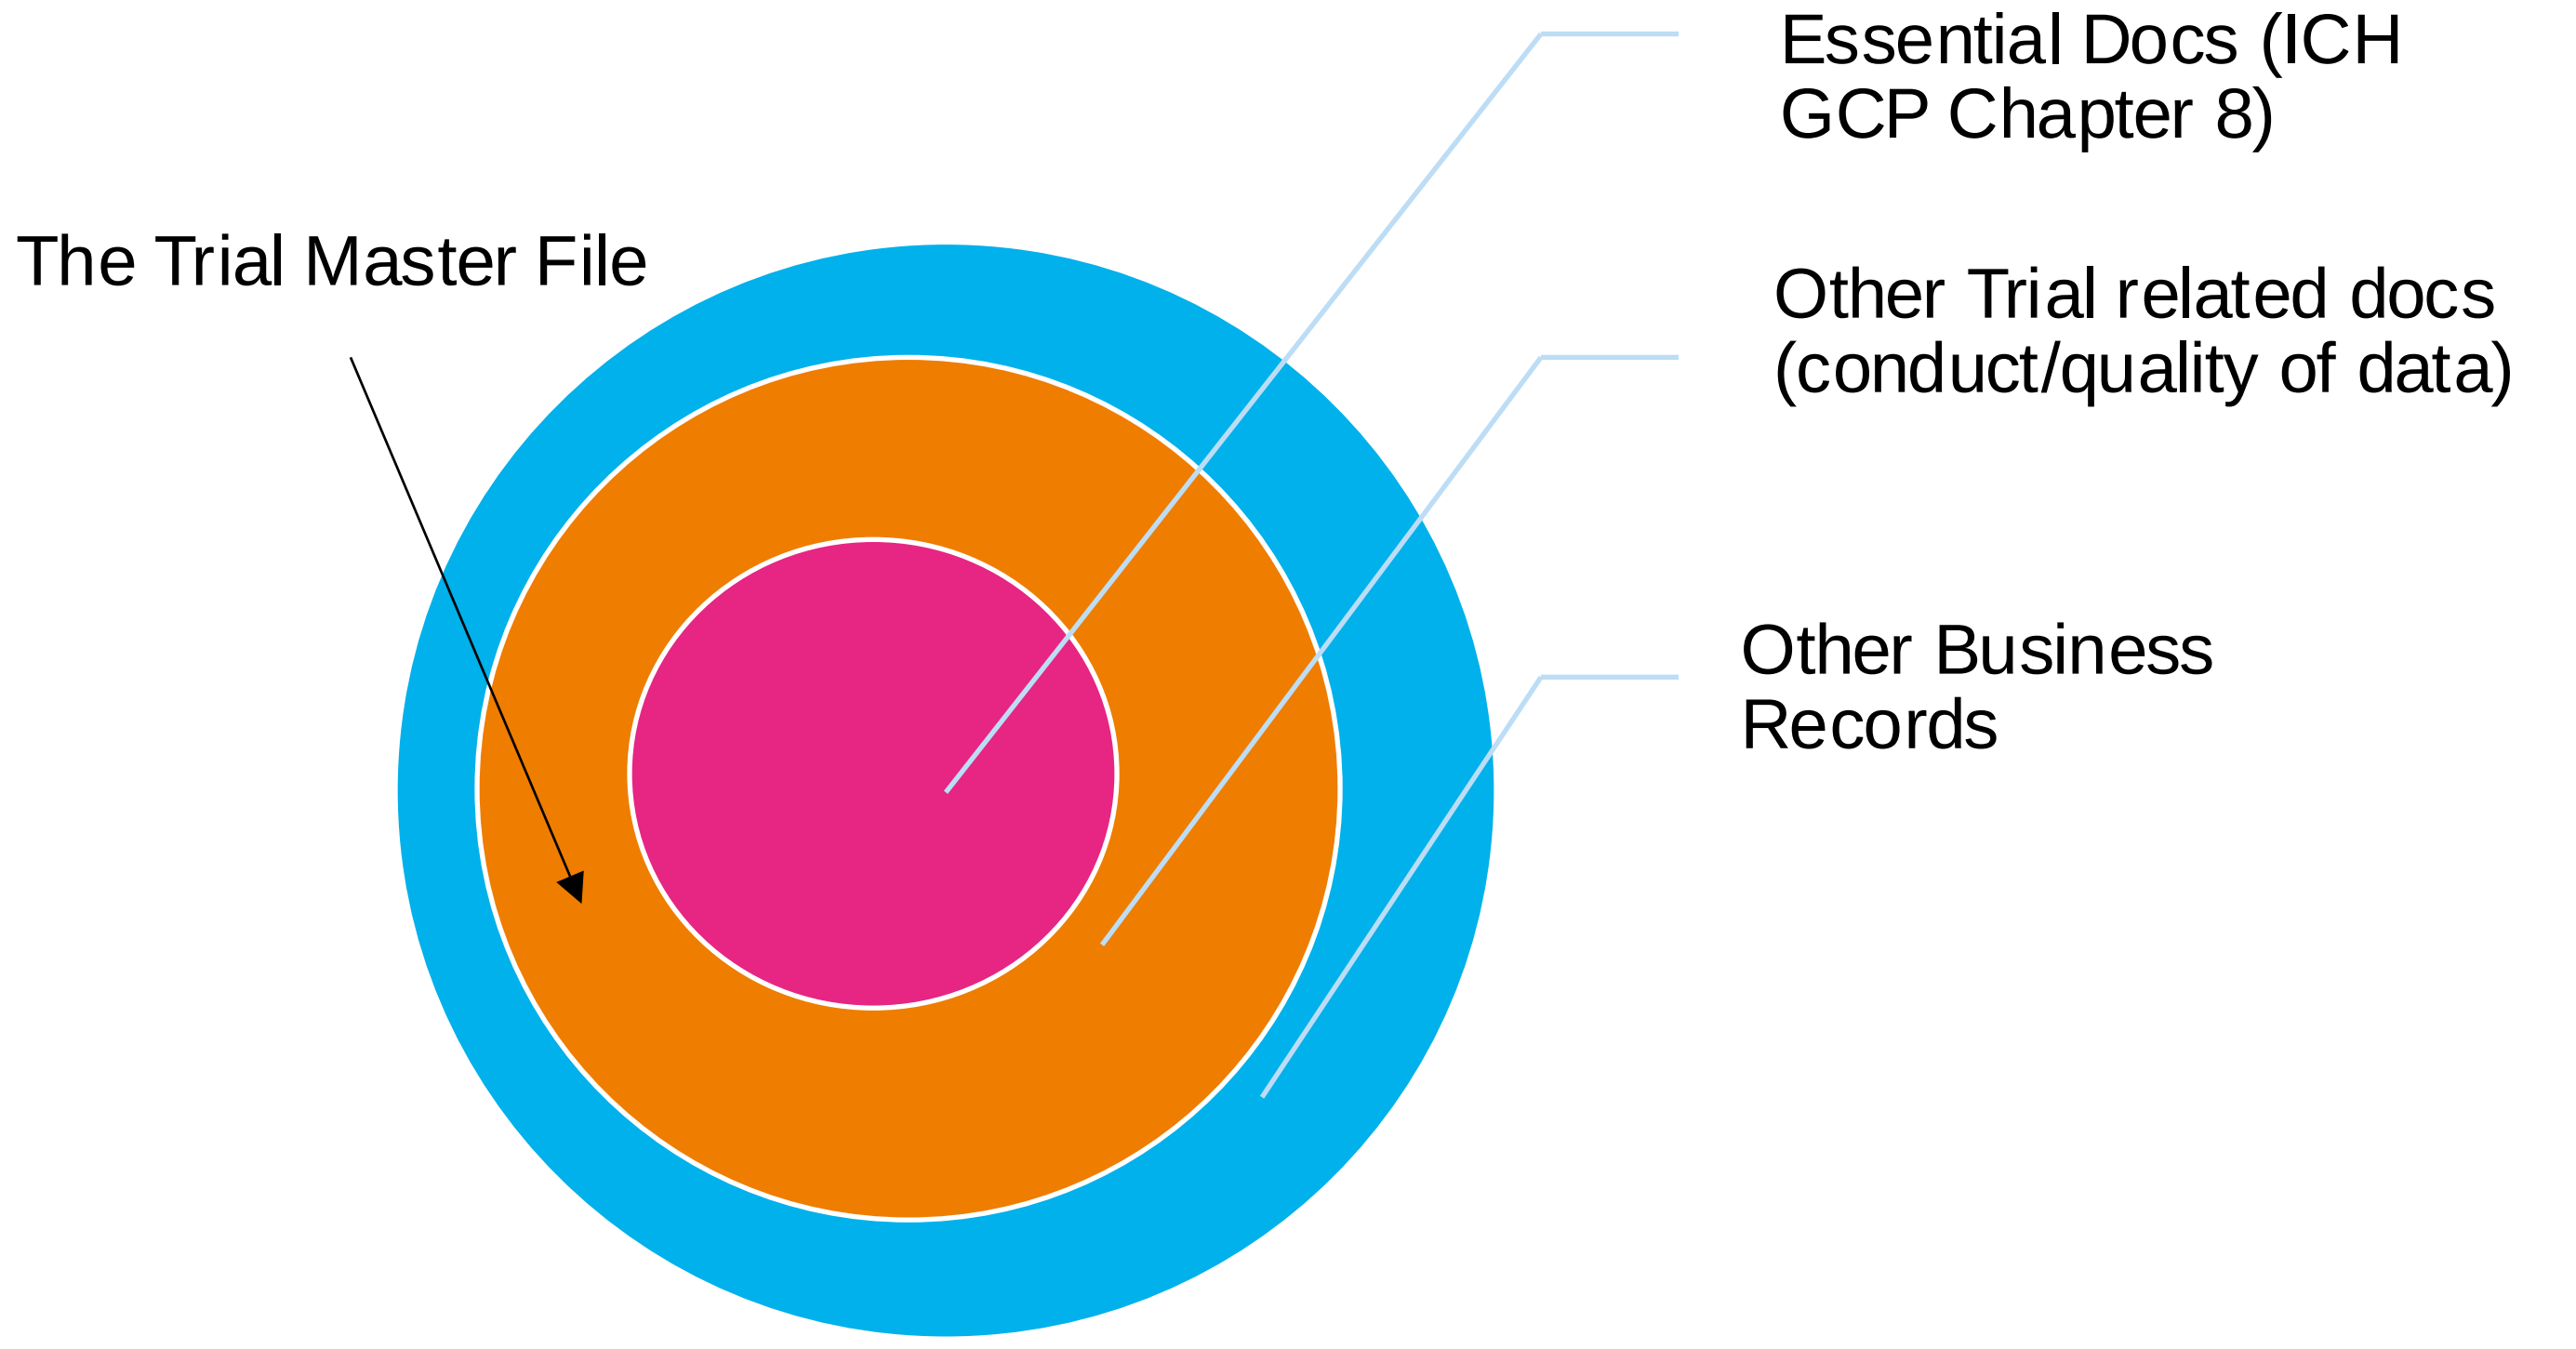
Essential Docs (ICH GCP Chapter 8)
The Trial Master File
Other Trial related docs (conduct/quality of data)
Other Business Records

Trial Master File Importance

Upon inspection, some common deficiencies that a clinical study sponsor, contract research organization (CRO), or monitor may experience in clinical studies include insufficient monitoring, failure to bring investigators into compliance, incomplete accountability for the investigational product, failure to appropriately document protocol deviations, inadequate subject protection including informed consent issues, record keeping issues, and failure to obtain IRB approval before study initiation activities. An accurate and complete eTMF decreases the likelihood of these types of documentation and regulatory compliance deficiencies.

Benefits of Trial Master File

The eTMF is an important output component of a clinical trial for both internal and external stakeholders. It contains and organizes documents such as the Final Report and Case Report Forms that are included in a PMTA and MRTPA (Modified Risk Tobacco Product Application). The eTMF for each study contains all the documents needed to allow an FDA inspector to reconstruct a clinical trial and answer an inspector's questions.

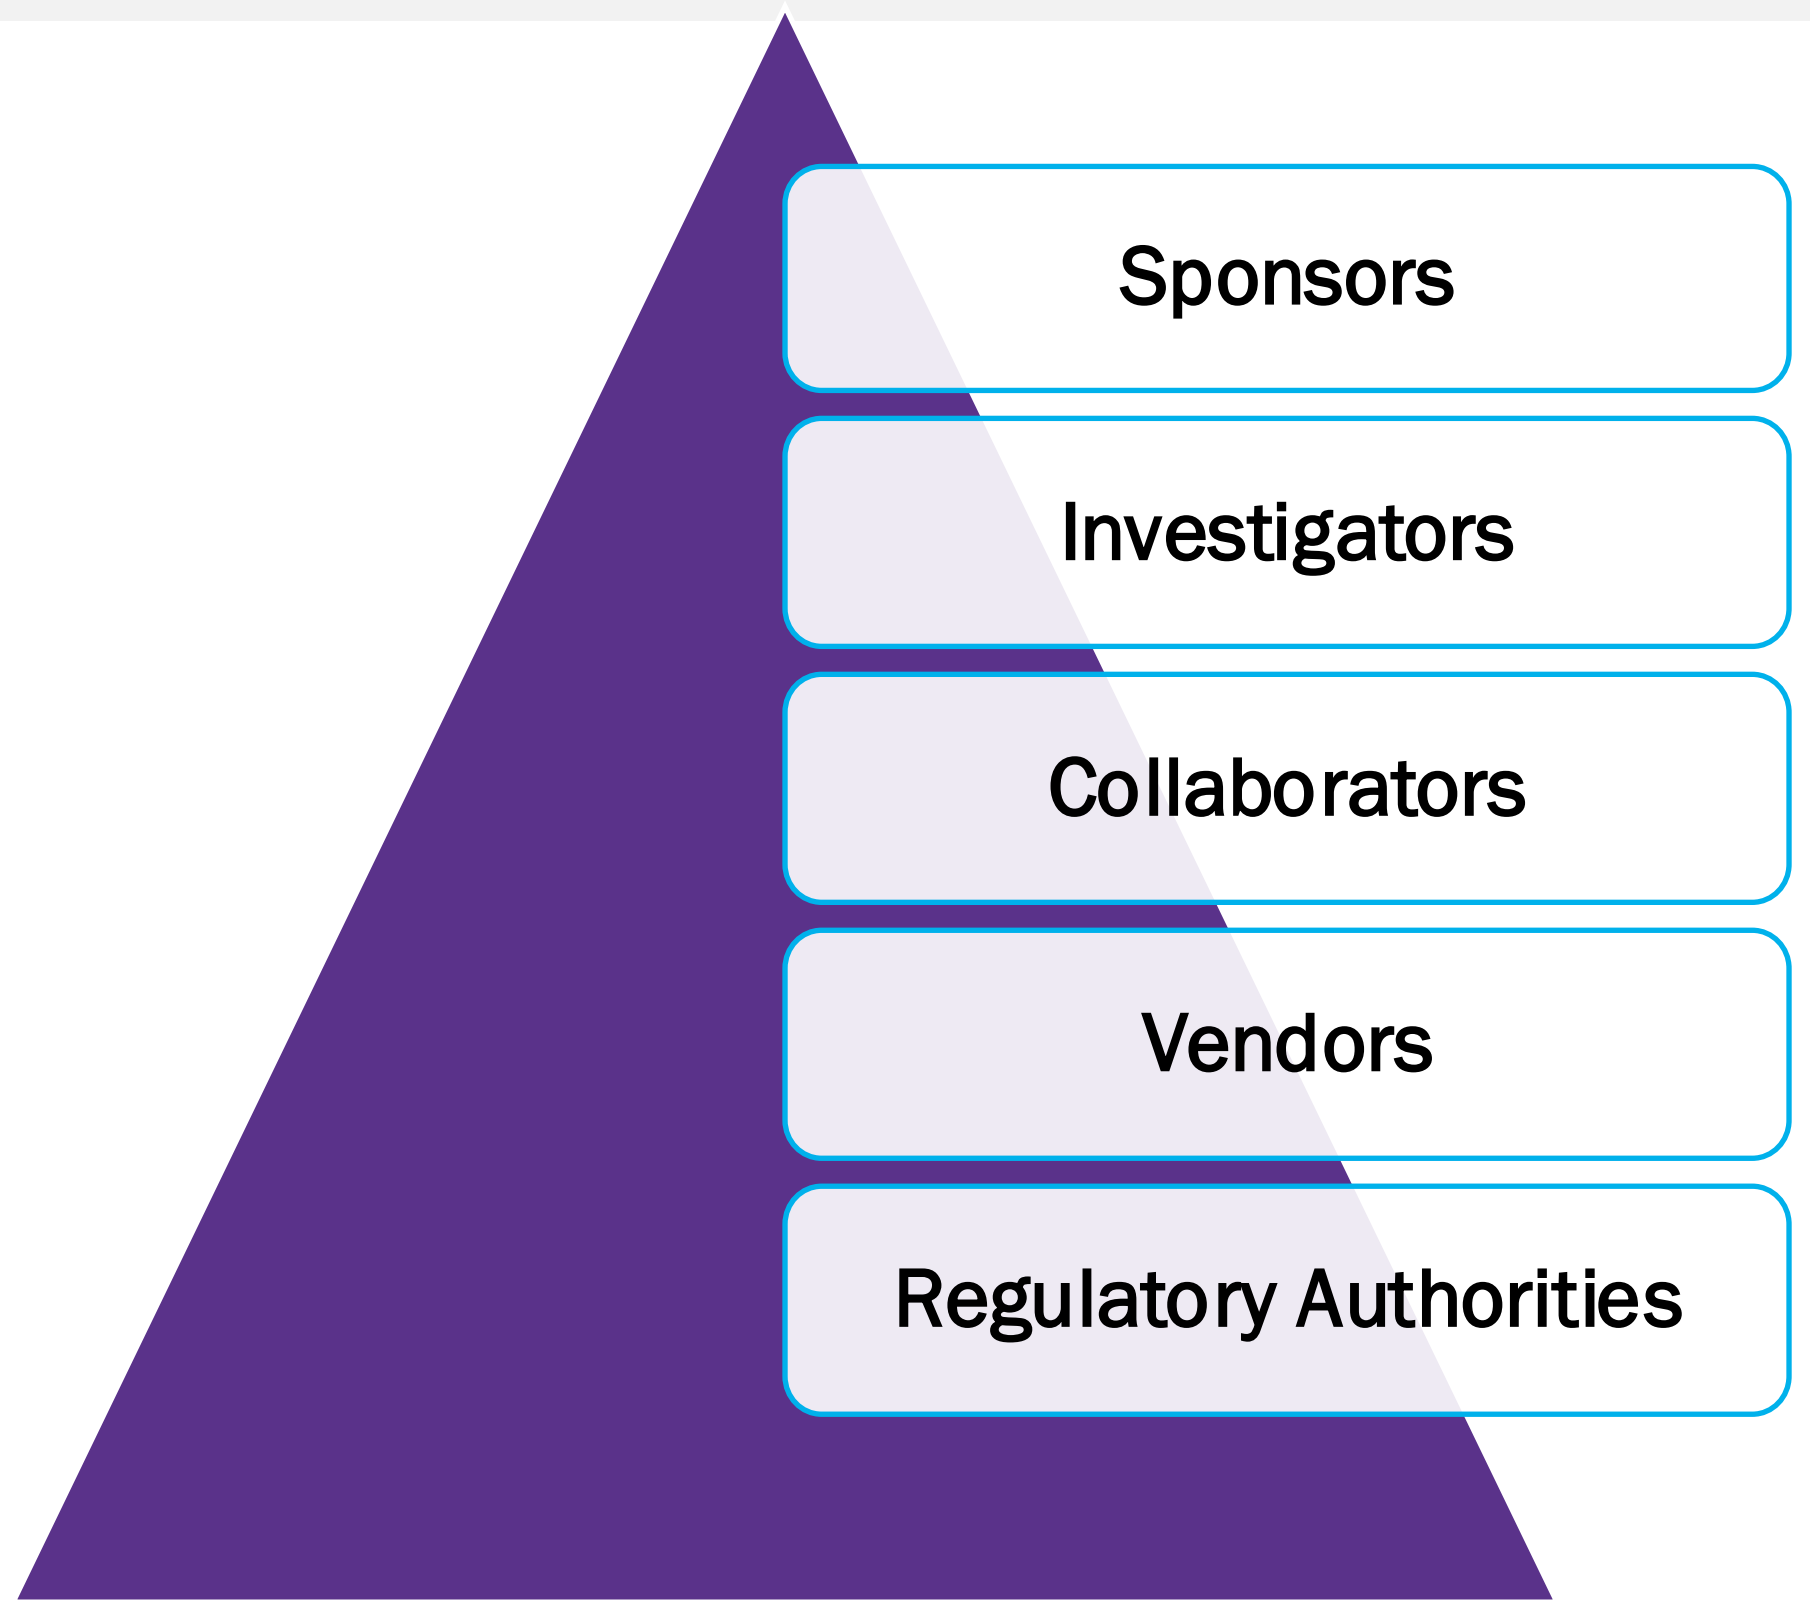
Sponsors
Investigators
Collaborators
Vendors
Regulatory Authorities

Summary

The eTMF helps clearly document how our company adheres to the rigorous quality standards for clinical research studies within the tobacco industry and has provided substantiation of clinical evidence supporting applications that have resulted in receipt of FDA Marketing Granted Orders for various new tobacco products. In conclusion, the efficacy of an archived eTMF is demonstrated in the retrieval-ready electronic documents available for regulatory applications, inspections, or review.

REYNOLDS
A BETTER TOMORROW

Contact: szeligk@rjrt.com

Reference: TMF University
https://www.lmkclinicalresearch.com/tmfu/

Point your phone's camera at the QR code to find our library of publications

Follow us:

www.ReynoldsScience.com

@RAI_News